isth 2022 CONGRESS JULY 9-13 ISTH2022.ORG LONDON

Effect of Novel Thrombolytic Aptamer BB-031 on Microfluidic Thrombolysis and VWF Function Dose-Response

Authors: R. Rassam[1], K. Thomas[2], C. Daniel[2], P. Spinella[1,3], S. Nimjee[4], S. Shea[1]

1. Trauma and Transfusion Medicine Research Center, Department of Surgery, University of Pittsburgh, Pittsburgh, PA
2. Department of Pediatrics, Washington University in St. Louis, St. Louis, MO
3. Department of Critical Care Medicine, University of Pittsburgh, Pittsburgh, PA
4. Department of Neurological Surgery, The Ohio State University Medical Center, Columbus, OH

University of Pittsburgh | Trauma and Transfusion Medicine Research Center
The Ohio State University Wexner Medical Center
Washington University in St. Louis

INTRODUCTION

The interaction between von Willebrand Factor (VWF) A1 domain and platelet GPIbα is essential for arterial thrombosis. Aptamer BB-031 selectively inhibits VWF A1 and has potent thrombolytic properties [1]. The ideal clinical assay to detect BB-031 functional efficacy and the corresponding therapeutic range are unknown. We measured dose responsiveness of BB-031 in a custom microfluidic model of thrombolysis and currently available clinical assays.

AIM

To determine the capacity of clinical assays to reflect microfluidic thrombolysis relative to measurable inhibition induced by BB-031.

METHOD

- Healthy human whole blood (N=10 unique donors) dosed with vehicle, 423nM, 846nM, 1692nM, or 3384nM BB-031 (*in vivo* dose range 0.25-2mg/kg)
- VWF function was assayed via VWF: Ag, VWF:Ac (Siemens GP1bα-interaction specific assay), T-TAS, VWF:Rco, and ristocetin impedance aggregometry (RIA)
- Microfluidic model of reperfusion:
 - WB was stained with fluorescent anti-CD41 conjugated to DyLight 350 (**PLT_{Throm}**) and perfused at high shear until upstream pressure increased to 25mm Hg
 - WB from the same donor stained with fluorescent anti-CD41 conjugated to Janelia Fluor 646 (**PLT_{Perf}**) dosed with vehicle, 1692nM, or 3385nM BB-031 and gently perfused to the site of thrombus
 - Dosed WB is left at a constant arterial pressure head and the site of thrombus is imaged for 120 minutes to quantify **PLT_{Throm}** and **PLT_{Perf}**
 - The max fold change in surface area (SA) of original and new deposition was quantified, and max fold change of **PLT_{Throm}** was used as microfluidic lytic efficacy (MLE) in correlation analysis

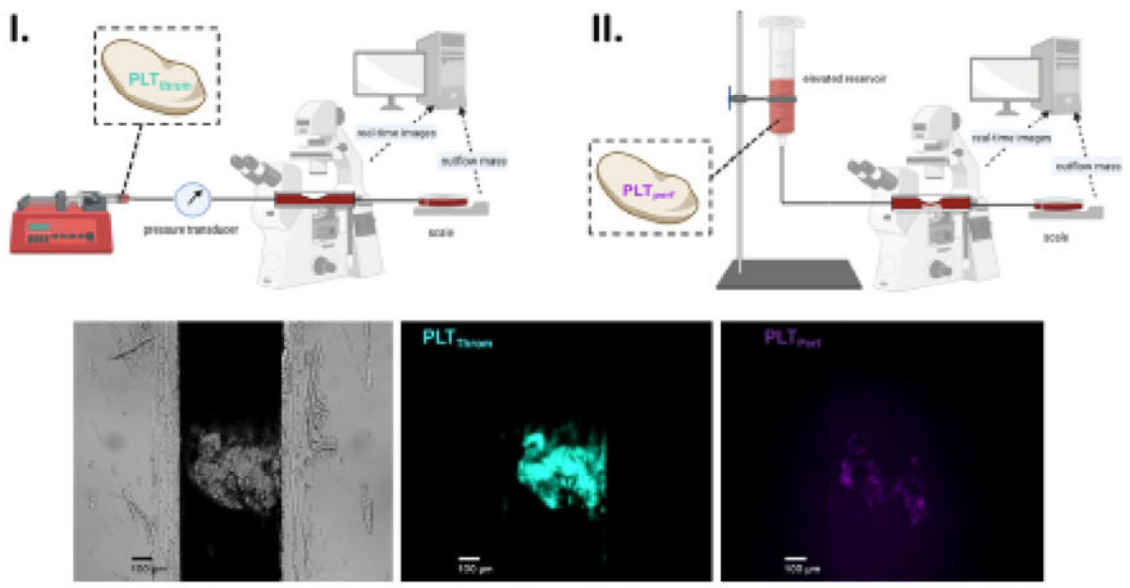

I.
II.
PLT Throm
PLT Perf
pressure transducer
real-time images
outflow mass
scale
elevated reservoir

Figure 1. Top: occlusion and post-occlusion perfusion approach. Bottom: a control thrombus immediately post-reperfusion.

RESULTS

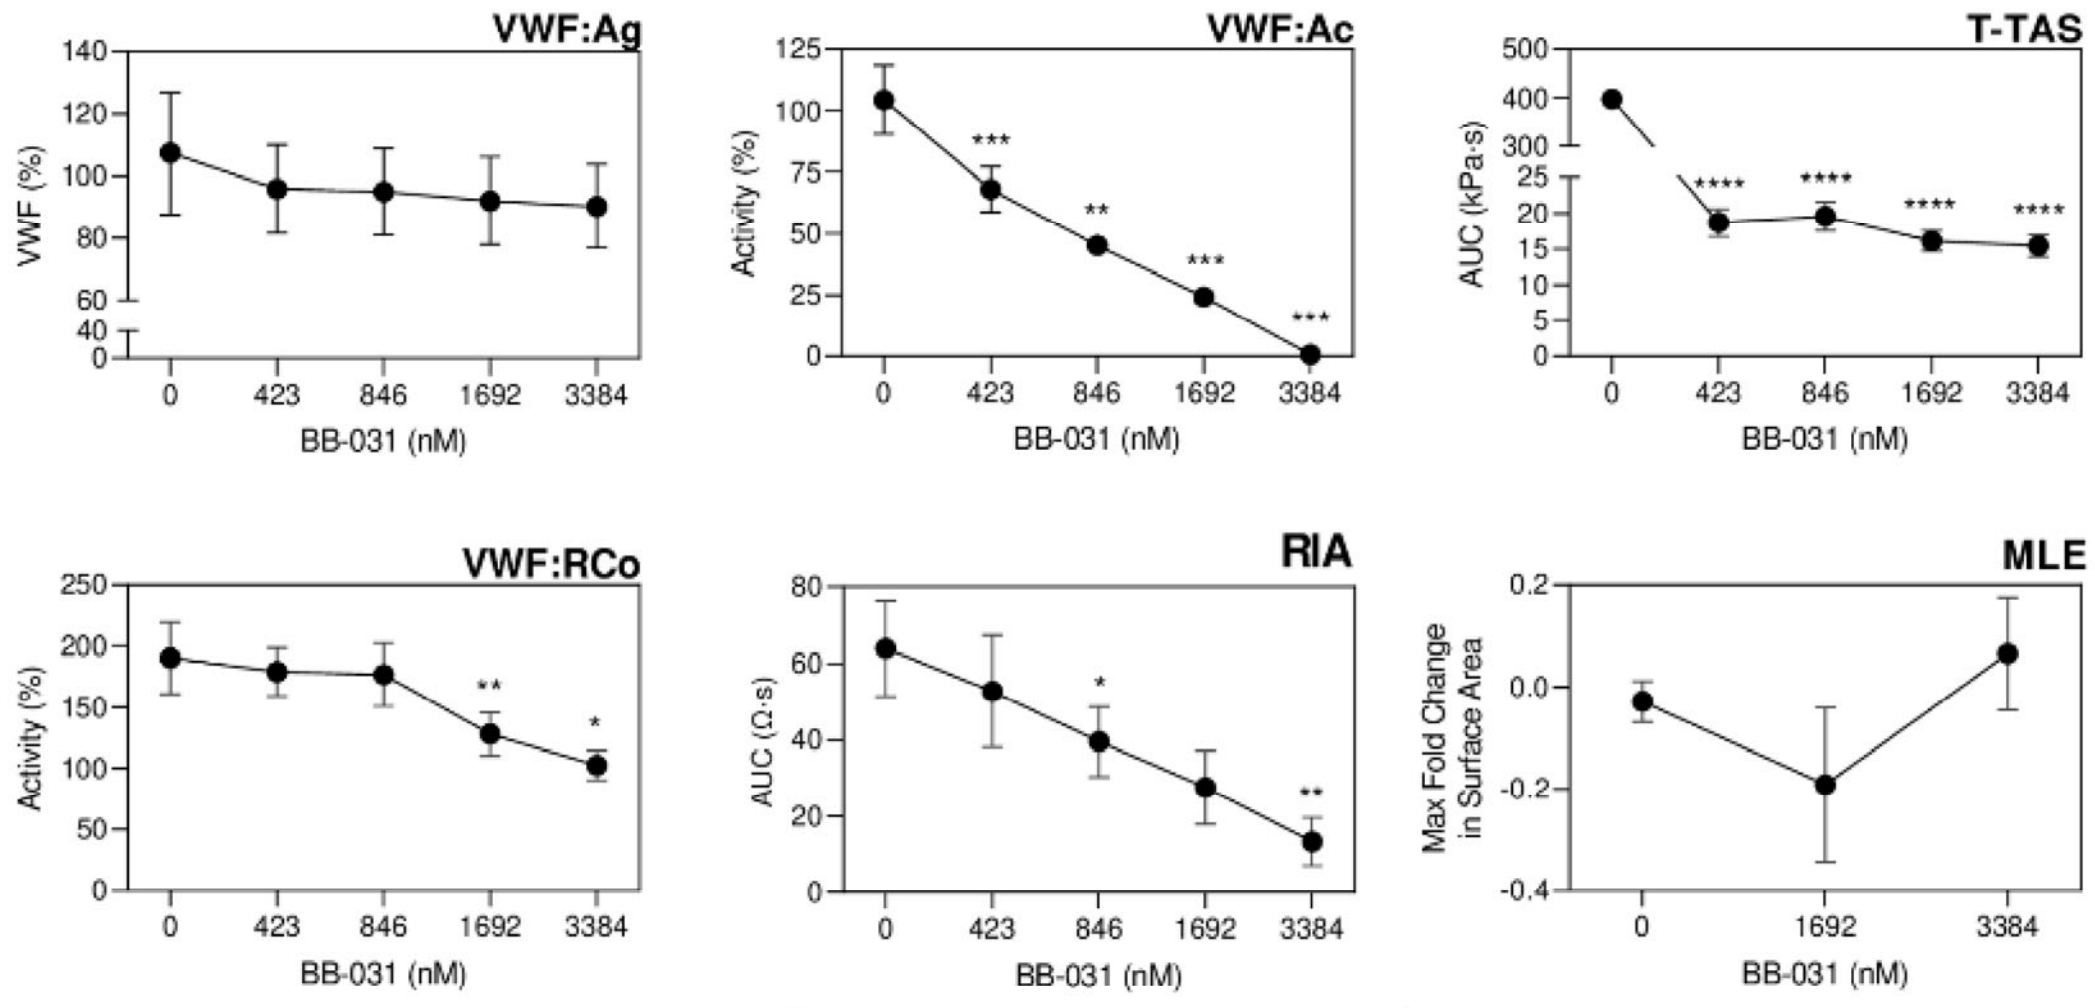

VWF:Ag
VWF (%)
VWF:Ac
Activity (%)
T-TAS
AUC (kPa·s)
VWF:RCo
Activity (%)
RIA
AUC (Ω·s)
MLE
Max Fold Change in Surface Area
BB-031 (nM)
0 423 846 1692 3384
* p<0.05, ** p<0.01, *** p<0.001, **** p<0.0001

Figure 2. VWF antigen levels and functional assays with increasing doses of BB-031. VWF:Ag: antigen; VWF:Ac: GP1bα-dependent activity; T-TAS: commercially available microfluidic device; VWF:RCo: ristocetin cofactor; RIA: ristocetin impedance aggregometry; MLE: microfluidic lytic efficacy, maximum fold change in surface area of original thrombus surface area. N=10 (same donor set) for all assays except MLE, which is performed on a subset of N=5. Data are displayed as summary points at the mean with error bars representing standard error of the mean (SEM).

VWF:Ag was not impacted by BB-031 dose and VWF functional assays were inhibited by BB-031 (Figure 2) in a dose-dependent fashion, as expected. Patency was achieved in 60% (3/5) of samples reperfused with 1692nM, 40% (2/5) of samples with 3384nM, and 0% (0/5) of samples perfused with vehicle. **PLT_{Throm}** SA changed by +1.8% (1.41%) (mean (SEM)) over 120min when perfused with vehicle, -17.20% (13.93%) when perfused with 1692nM BB-031 (p=0.096 vs. vehicle), and -5.17% (7.41%) when perfused with 3384nM BB-031 (p=0.482 vs. vehicle, p=0.0134 vs. 1692nM). SA of **PLT_{Perf}** maximally increased by 10.76% (6.68%) when perfused with vehicle, 30.23% (21.83%) when perfused with 1692nM BB-031 (p=0.028 vs. vehicle), and 65.81% (36.73%) when perfused 3384nM BB-031 (p=0.067 vs. vehicle, p=0.002 vs. 1692nM). However, PLTperf completely lysed away by 120min with BB-031 reperfusion.

To further investigate the point at which function on T-TAS is inhibited by BB-031, an additional dose response was performed only for this assay (Figure 3).

VWF:Ac, VWF:Rco, and RIA correlated best with BB-031 dose. MLE correlated best with T-TAS, a measure of function under flow (Figure 4).

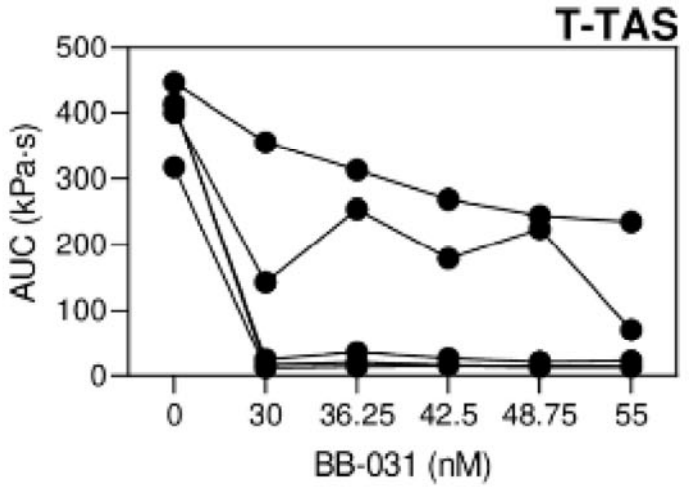

T-TAS
AUC (kPa·s)
BB-031 (nM)
0 30 36.25 42.5 48.75 55

Figure 4. Lower doses of BB-031 on T-TAS in a subset of 5 donors.

	Dose	VWF:Ag	VWF:Ac	T-TAS	VWF:RCo	RIA	MLE
Dose	1.00	-0.77	-0.92	-0.54	-0.97	-0.94	0.47
VWF:Ag	-0.77	1.00	0.94	0.95	0.77	0.87	-0.79
VWF:Ac	-0.92	0.94	1.00	0.79	0.91	0.98	-0.71
T-TAS	-0.54	0.95	0.79	1.00	0.52	0.67	-0.85
VWF:RCo	-0.97	0.77	0.91	0.52	1.00	0.94	-0.66
RIA	-0.94	0.87	0.98	0.67	0.94	1.00	-0.72
MLE	0.47	-0.79	-0.71	-0.85	-0.66	-0.72	1.00

Colour scale: 1.0, 0.5, 0, -0.5, -1.0

Figure 3. Correlation matrix among all assays.

CONCLUSIONS

BB-031 treatment inhibited VWF function and was successful in reducing thrombus SA of the original occlusive thrombi and thrombi deposited during reperfusion, and in achieving recanalization.

Surprisingly, reperfusion with 3384nM BB-031 was less efficacious than 1692nM both in thrombolysis and inhibition of new deposition, perhaps due to off-target effects.

While VWF:RCo had the highest correlation coefficient with dose, VWF:Ac was maximally inhibited at the highest dose and still correlated very well with dose. It may also be a more logistically feasible clinical assay though is not yet FDA-approved in the USA.

Continued study of BB-031 is underway to investigate effects of thrombus retraction post-occlusion on lysis. A novel system is being developed to achieve complete occlusion with drug delivery just upstream of the thrombus without disruption.

ACKNOWLEDGEMENTS

The authors would like to thank Dr. David Lillicrap, Christine Brown, and Colleen Notley (Department of Pathology and Molecular Medicine, Queen's University, Kingston, Ontario) for prompt correspondence, receipt, processing, and data delivery regarding the VWF:Ac assay. The authors would also like to thank Dr. David Ku (School of Mechanical Engineering, Georgia Institute of Technology, Atlanta, GA), Dr. Michael Griffin, and Dr. Lauren Casa for design and supply of the microfluidic chamber.

This study was funded by Basking Biosciences via a Collaboration Agreement with Washington University in St. Louis. Continued work is funded by Basking Biosciences at the University of Pittsburgh.

REFERENCES

1. Nimjee SM, Dornbos D, Pitoc GA, Wheeler DG, Layzer JM, Venetos N, Huttinger A, Talentino SE, Musgrave NJ, Moody H, Rempel RE, Jones C, Carlisle K, Wilson J, Bratton C, Joseph ME, Khan S, Hoffman MR, Sommerville L, Becker RC, Zweier JL, Sullenger BA. Preclinical Development of a vWF Aptamer to Limit Thrombosis and Engender Arterial Recanalization of Occluded Vessels. Mol Ther. 2019; 27: 1228-41. 10.1016/j.ymthe.2019.03.016.

CONTACT INFORMATION

Susan M. Shea, PhD
Assistant Professor
Department of Surgery
University of Pittsburgh
Pittsburgh, PA, USA

smshea@pitt.edu
@SMSheaLab